Abstract
4155

Regulation of MDSC trafficking and function in RCC by CXCR4 in the presence of a VEGF-R antagonist

David J Panka[1], Robert D Arbeit[2] and James W Mier[1]

[1]Beth Israel Deaconess Medical Center, Boston, MA and [2]X4 Pharmaceuticals Inc, Cambridge, MA

Introduction

Using a murine model of 786-0 and A498 RCC xenografts, we have previously demonstrated that acquired resistance to sunitinib treatment was associated with a marked increase in the infiltration of CD11b$^+$/Gr-1$^+$ myeloid-derived suppressor cells (MDSC). These cells have also been implicated in the development of resistance to other anticancer therapies. Further, we observed that both the influx of MDSC and resistance to VEGF-targeted therapies could be prevented by concurrent administration of an HDM2 antagonist, a drug whose biological effects are mediated primarily through the up regulation of p53. MDSC trafficking into tumor tissue is regulated by chemokines, many of which (e.g. SDF-1/CXCL-12) are produced in response to HIF-2 expression. p53 is known to directly repress CXCL12 transcription, and we have shown that HDM2 blockade suppresses HIF-2 expression, suggesting that the drug has both direct and indirect effects on CXCL12 expression. Western blot analysis of tumor lysates confirmed that HDM2 antagonism mediates its effects on MDSC through the suppression of chemokine production, including CXCL12. These findings suggested that the ability of HDM2 antagonism to prevent sunitinib resistance might be due, at least in part, to the suppression of CXCL12 production and MDSC recruitment. Consequently we hypothesized that agents that block CXCL12/CXCR4 signaling directly would duplicate the effects of HDM2 blockade on MDSC trafficking and prevent resistance to VEGF-targeted therapies.

Methods

Mice were inoculated with 786-0 and A498 RCC xenografts, the tumors permitted to grow to ~300 mm^3, and then treatment initiated with the CXCR4 inhibitor X4P-001, axitinib, both agents in combination, or saline (control). Tumors were treated and measured daily for 22 days. Tumors were removed and either snap frozen in liquid nitrogen for western blot analysis or fixed in formalin for immunohistochemistry (Ki67) and immunofluorescence (MDSC; cd11b/Gr-1). IHC and IF analysis was quantitated using ImageJ software.

Summary and Conclusions

1. The combination treatment of X4P-001 and axitinib demonstrated significantly more potent anti-tumor activity than either single agent alone in two renal xenograft models

2. The addition of axitinib to RCC xenografts results in a significant increase in proliferation of tumors as measured by Ki67 staining. The increase of Ki67 staining is suppressed with the addition of X4P-001 to axitinib.

3. X4P-001 suppressed the increased MDSC tumor infiltration caused by axitinib treatment

4. Activities of several key signaling molecules including p-STAT3 and p-AKT were inhibited, consistent with the observed suppression of MDSC infiltration and tumor cell survival

5. Axitinib inhibited the activity of p53 in RCC xenografts as measured by p21 activity, which was unaffected by the addition of X4P-001 .

6. Data presented here support the rationale for the current clinical investigation of X4P-001 in RCC where suppressive TME is driven by hypoxia induced MDSCs (NCT02667886)

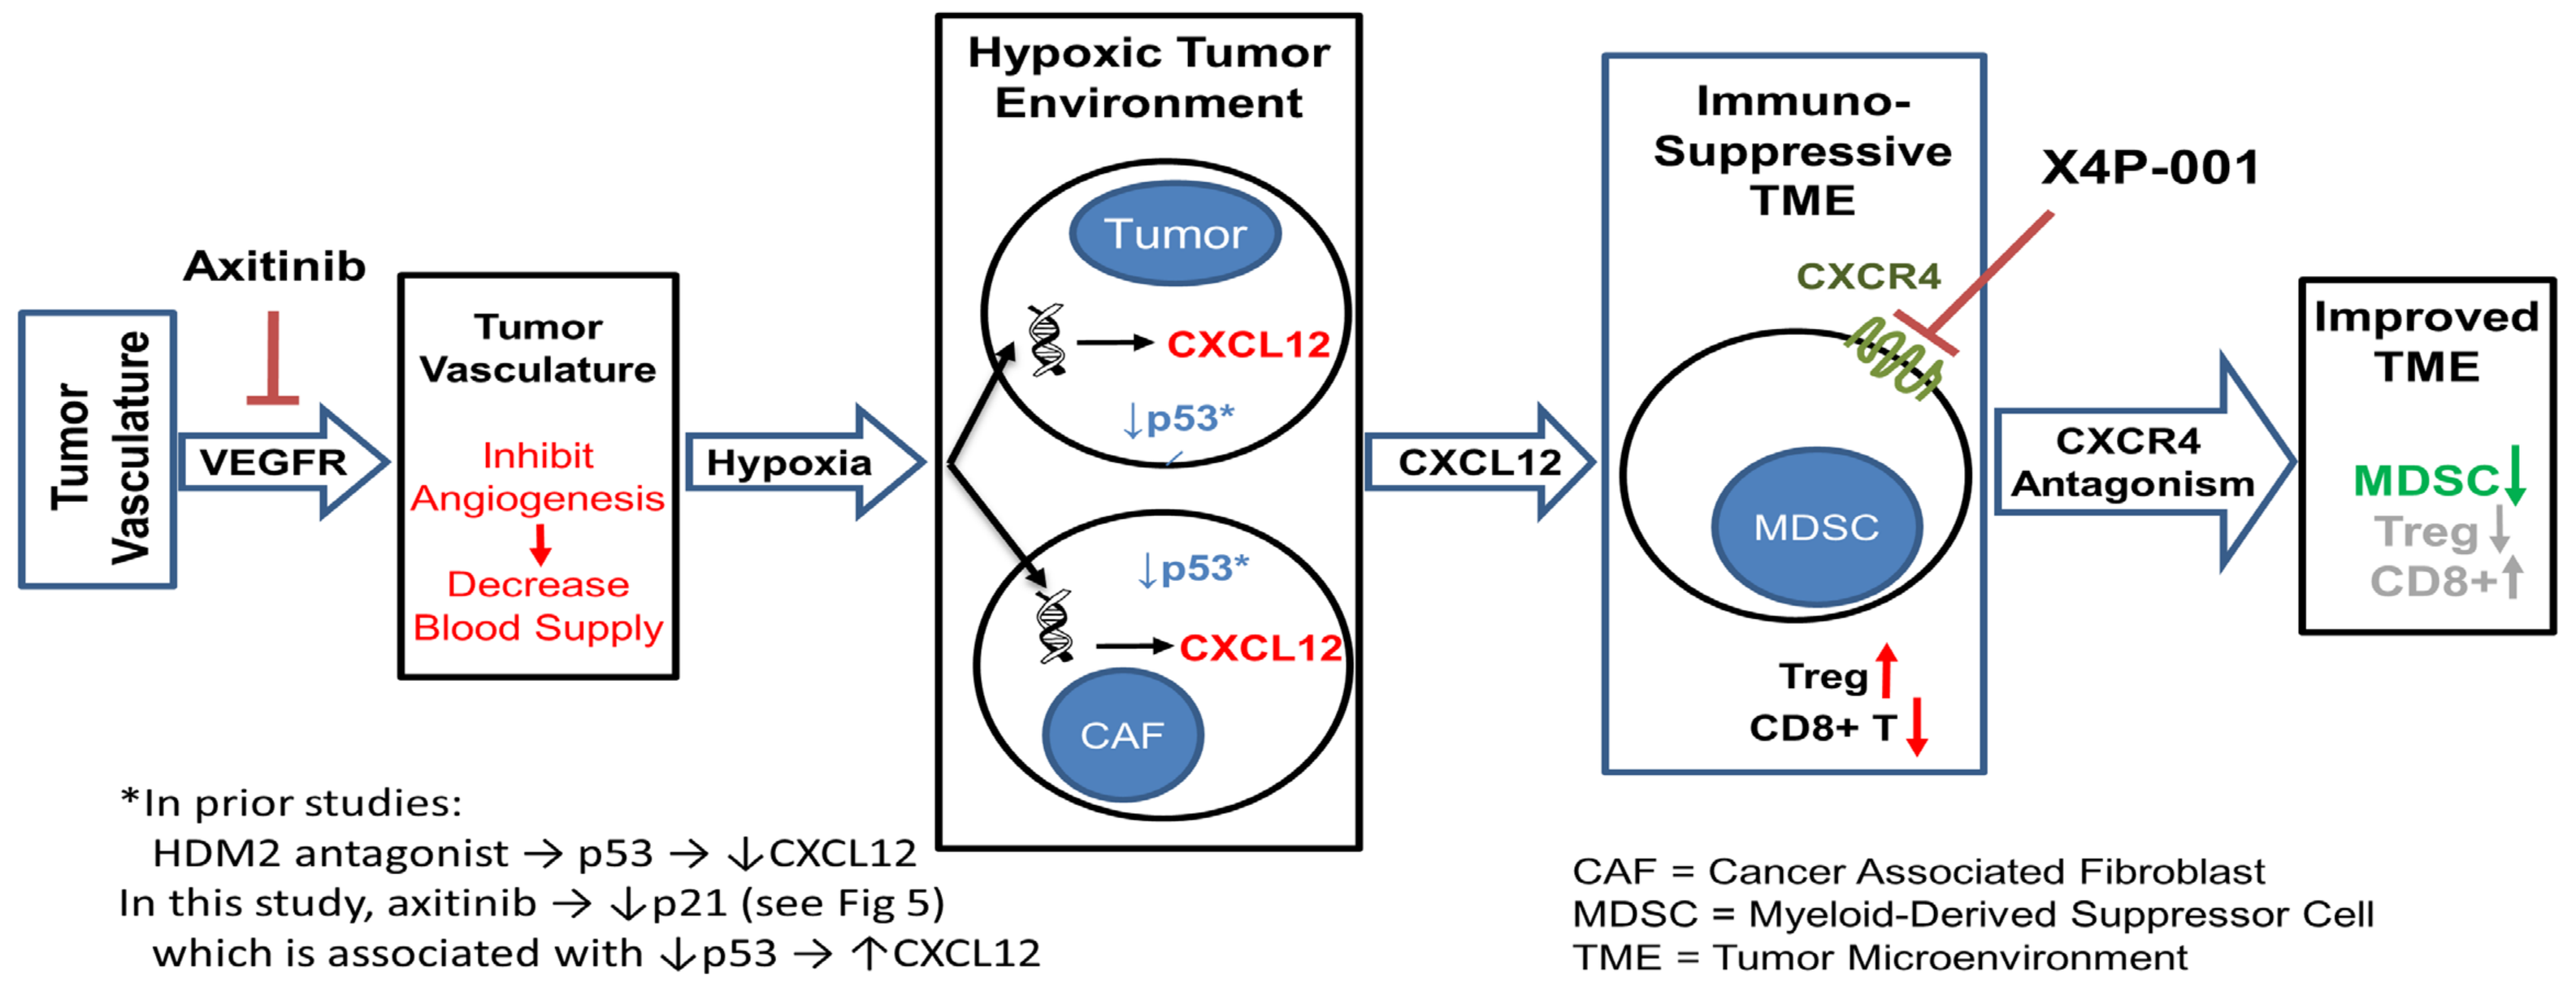
Axitinib
Tumor Vasculature
VEGFR
Tumor Vasculature
Inhibit Angiogenesis
Decrease Blood Supply
Hypoxia
Hypoxic Tumor Environment
Tumor
CXCL12
↓p53*
↓p53*
CXCL12
CAF
CXCL12
Immuno-Suppressive TME
CXCR4
X4P-001
MDSC
Treg↑
CD8+ T↓
CXCR4 Antagonism
Improved TME
MDSC↓
Treg↓
CD8+↑
*In prior studies:
HDM2 antagonist → p53 → ↓CXCL12
In this study, axitinib → ↓p21 (see Fig 5)
which is associated with ↓p53 → ↑CXCL12
CAF = Cancer Associated Fibroblast
MDSC = Myeloid-Derived Suppressor Cell
TME = Tumor Microenvironment

The combination of axitinib and X4P-001 retards tumor growth to a greater extent than either drug alone

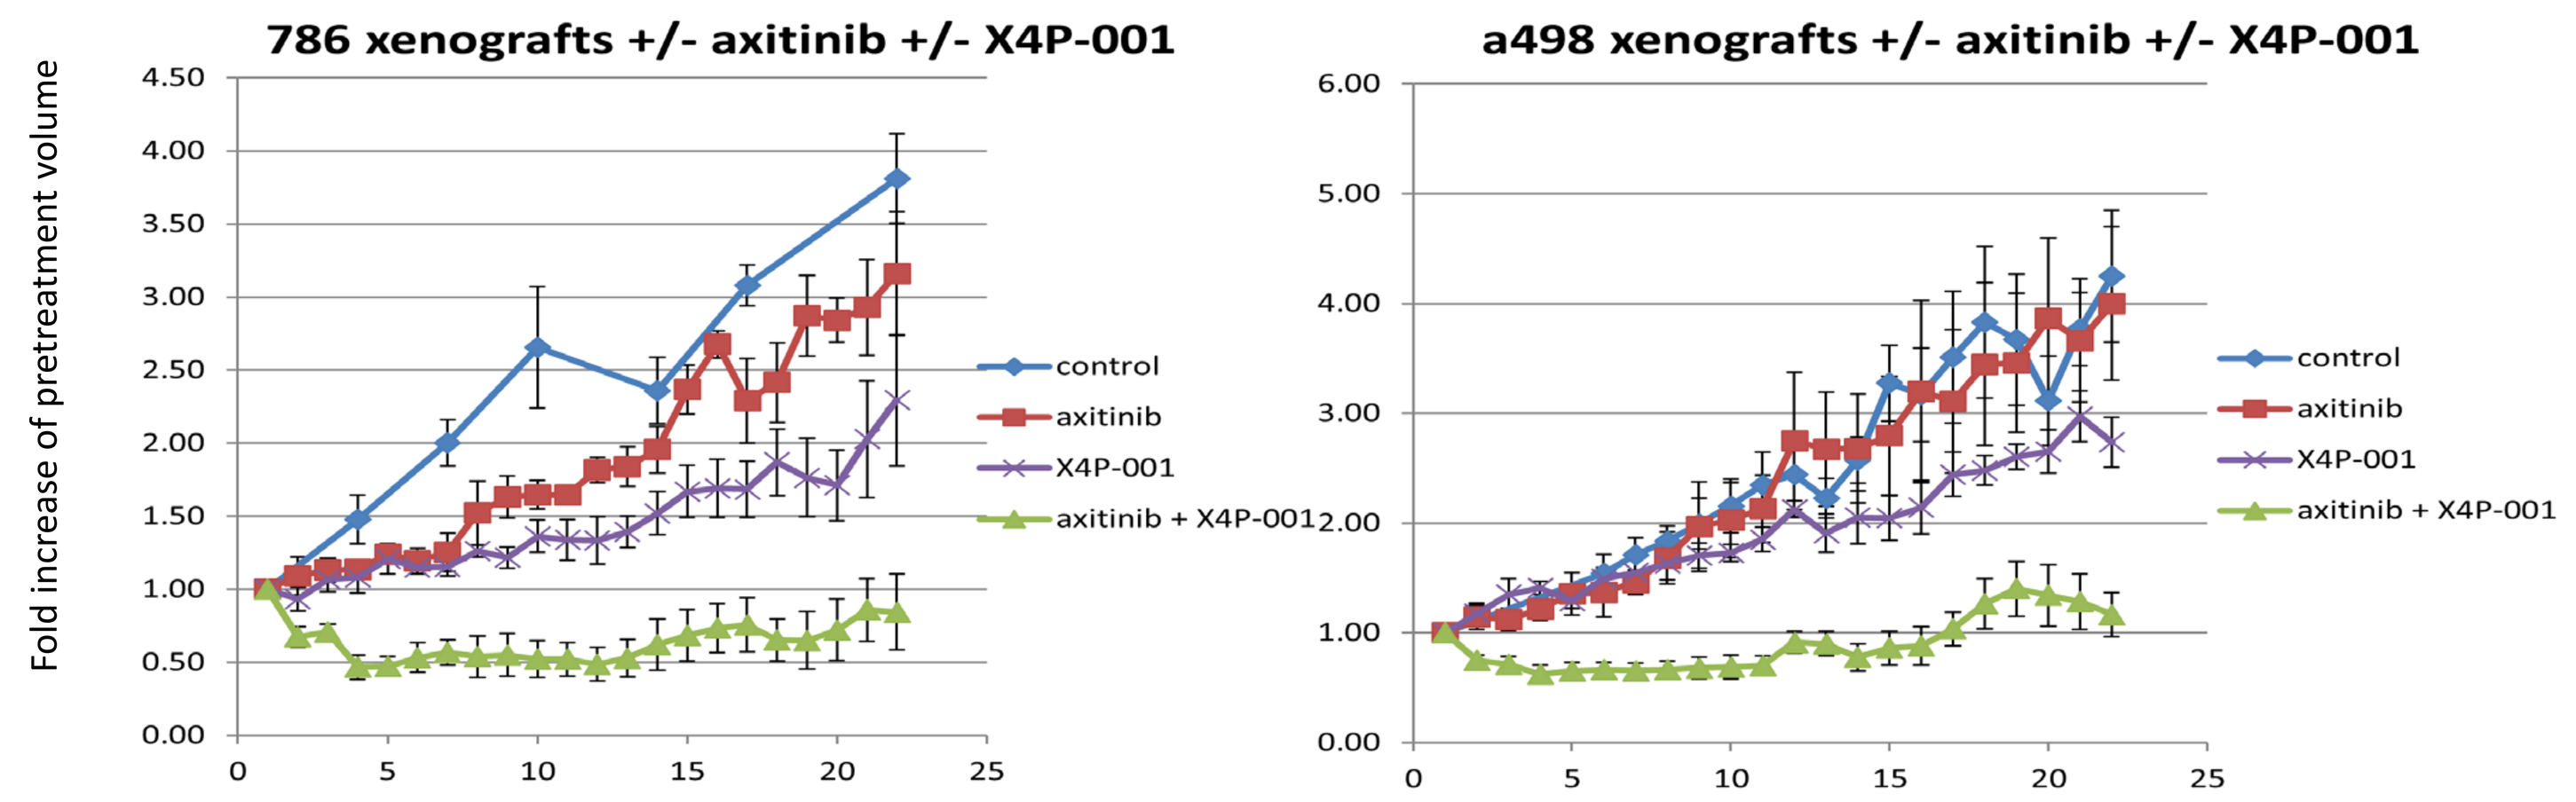
786 xenografts +/- axitinib +/- X4P-001
Fold increase of pretreatment volume
a498 xenografts +/- axitinib +/- X4P-001
control
axitinib
X4P-001
axitinib + X4P-001

Figure 1. Effect of X4P-001 on axitinib efficacy in 786-0 and A498 xenografts.

The combination of axitinib and X4P-001 results in more necrosis than either drug alone

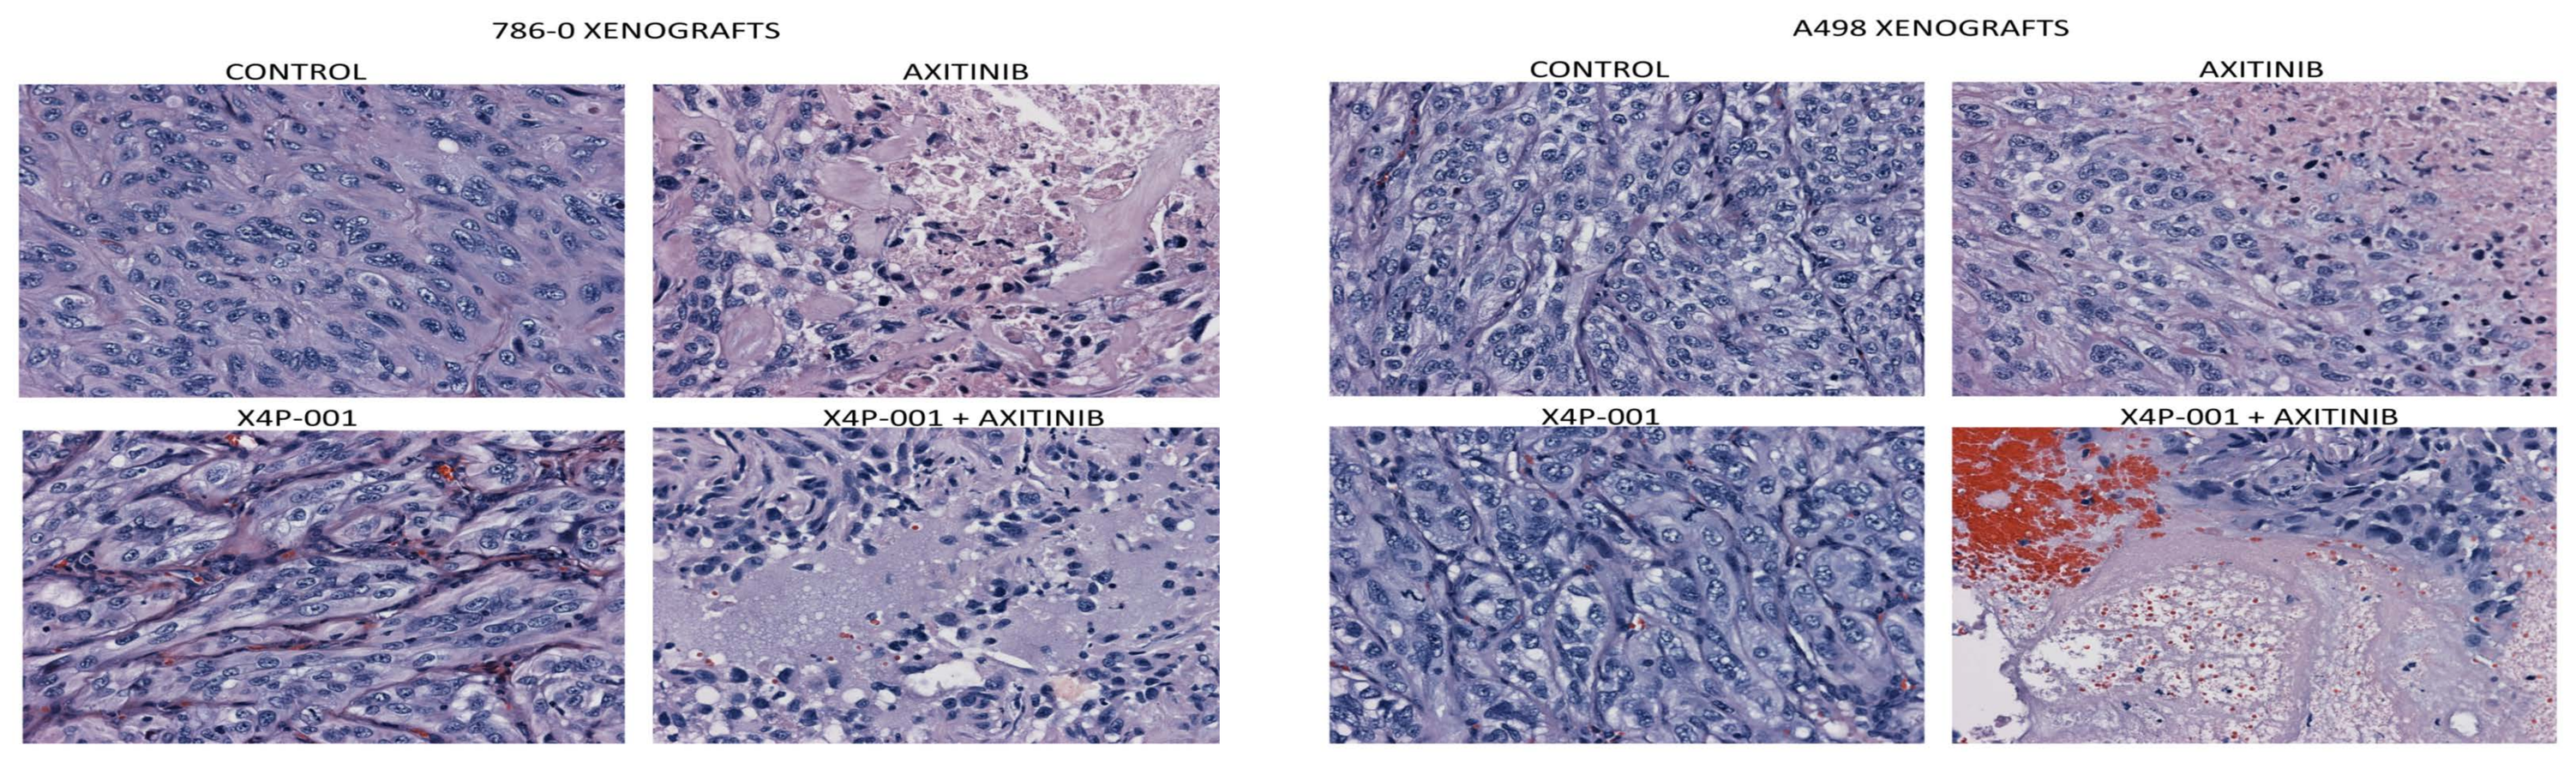
786-0 XENOGRAFTS
CONTROL
AXITINIB
X4P-001
X4P-001 + AXITINIB
A498 XENOGRAFTS
CONTROL
AXITINIB
X4P-001
X4P-001 + AXITINIB

Figure 2. Hemotoxylin and Eosin staining of 786 and A498 xenograft tumors treated with axitinib +/- X4P-001 taken at sacrifice (day 22).

The addition of axitinib to RCC xenografts results in a significant increase in proliferation of tumors at measure by Ki67 staining. The increase of Ki67 staining is suppressed with the addition of X4P-001 to axitinib.

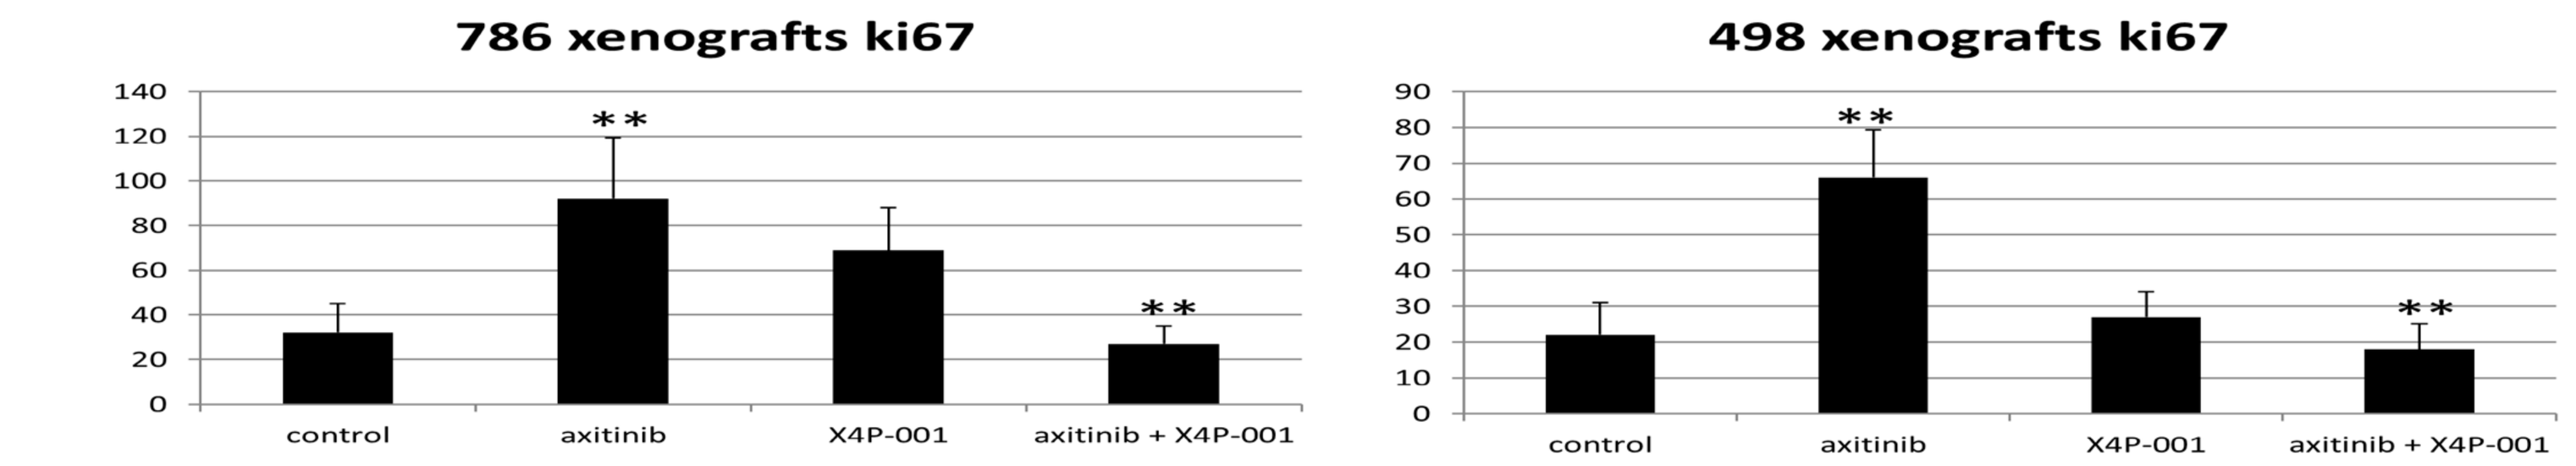
786 xenografts ki67
498 xenografts ki67
control
axitinib
X4P-001
axitinib + X4P-001

Figure 3. Immunohistochemistry of 786 and A498 xenograft tumors treated with axitinib +/- X4P-001 taken at sacrifice (day 22). Tumors were stained for Ki67. Data is presented as bar graphs of quantitative analysis of all tumors in each treatment group. (**p<0.05).

The addition of axitinib to RCC xenografts results in a significant increase in tumor MDSC infiltration as measured by cd11b and Gr-1 staining. The infiltration of MDSCs is suppressed with the addition of X4P-001 to axitinib.

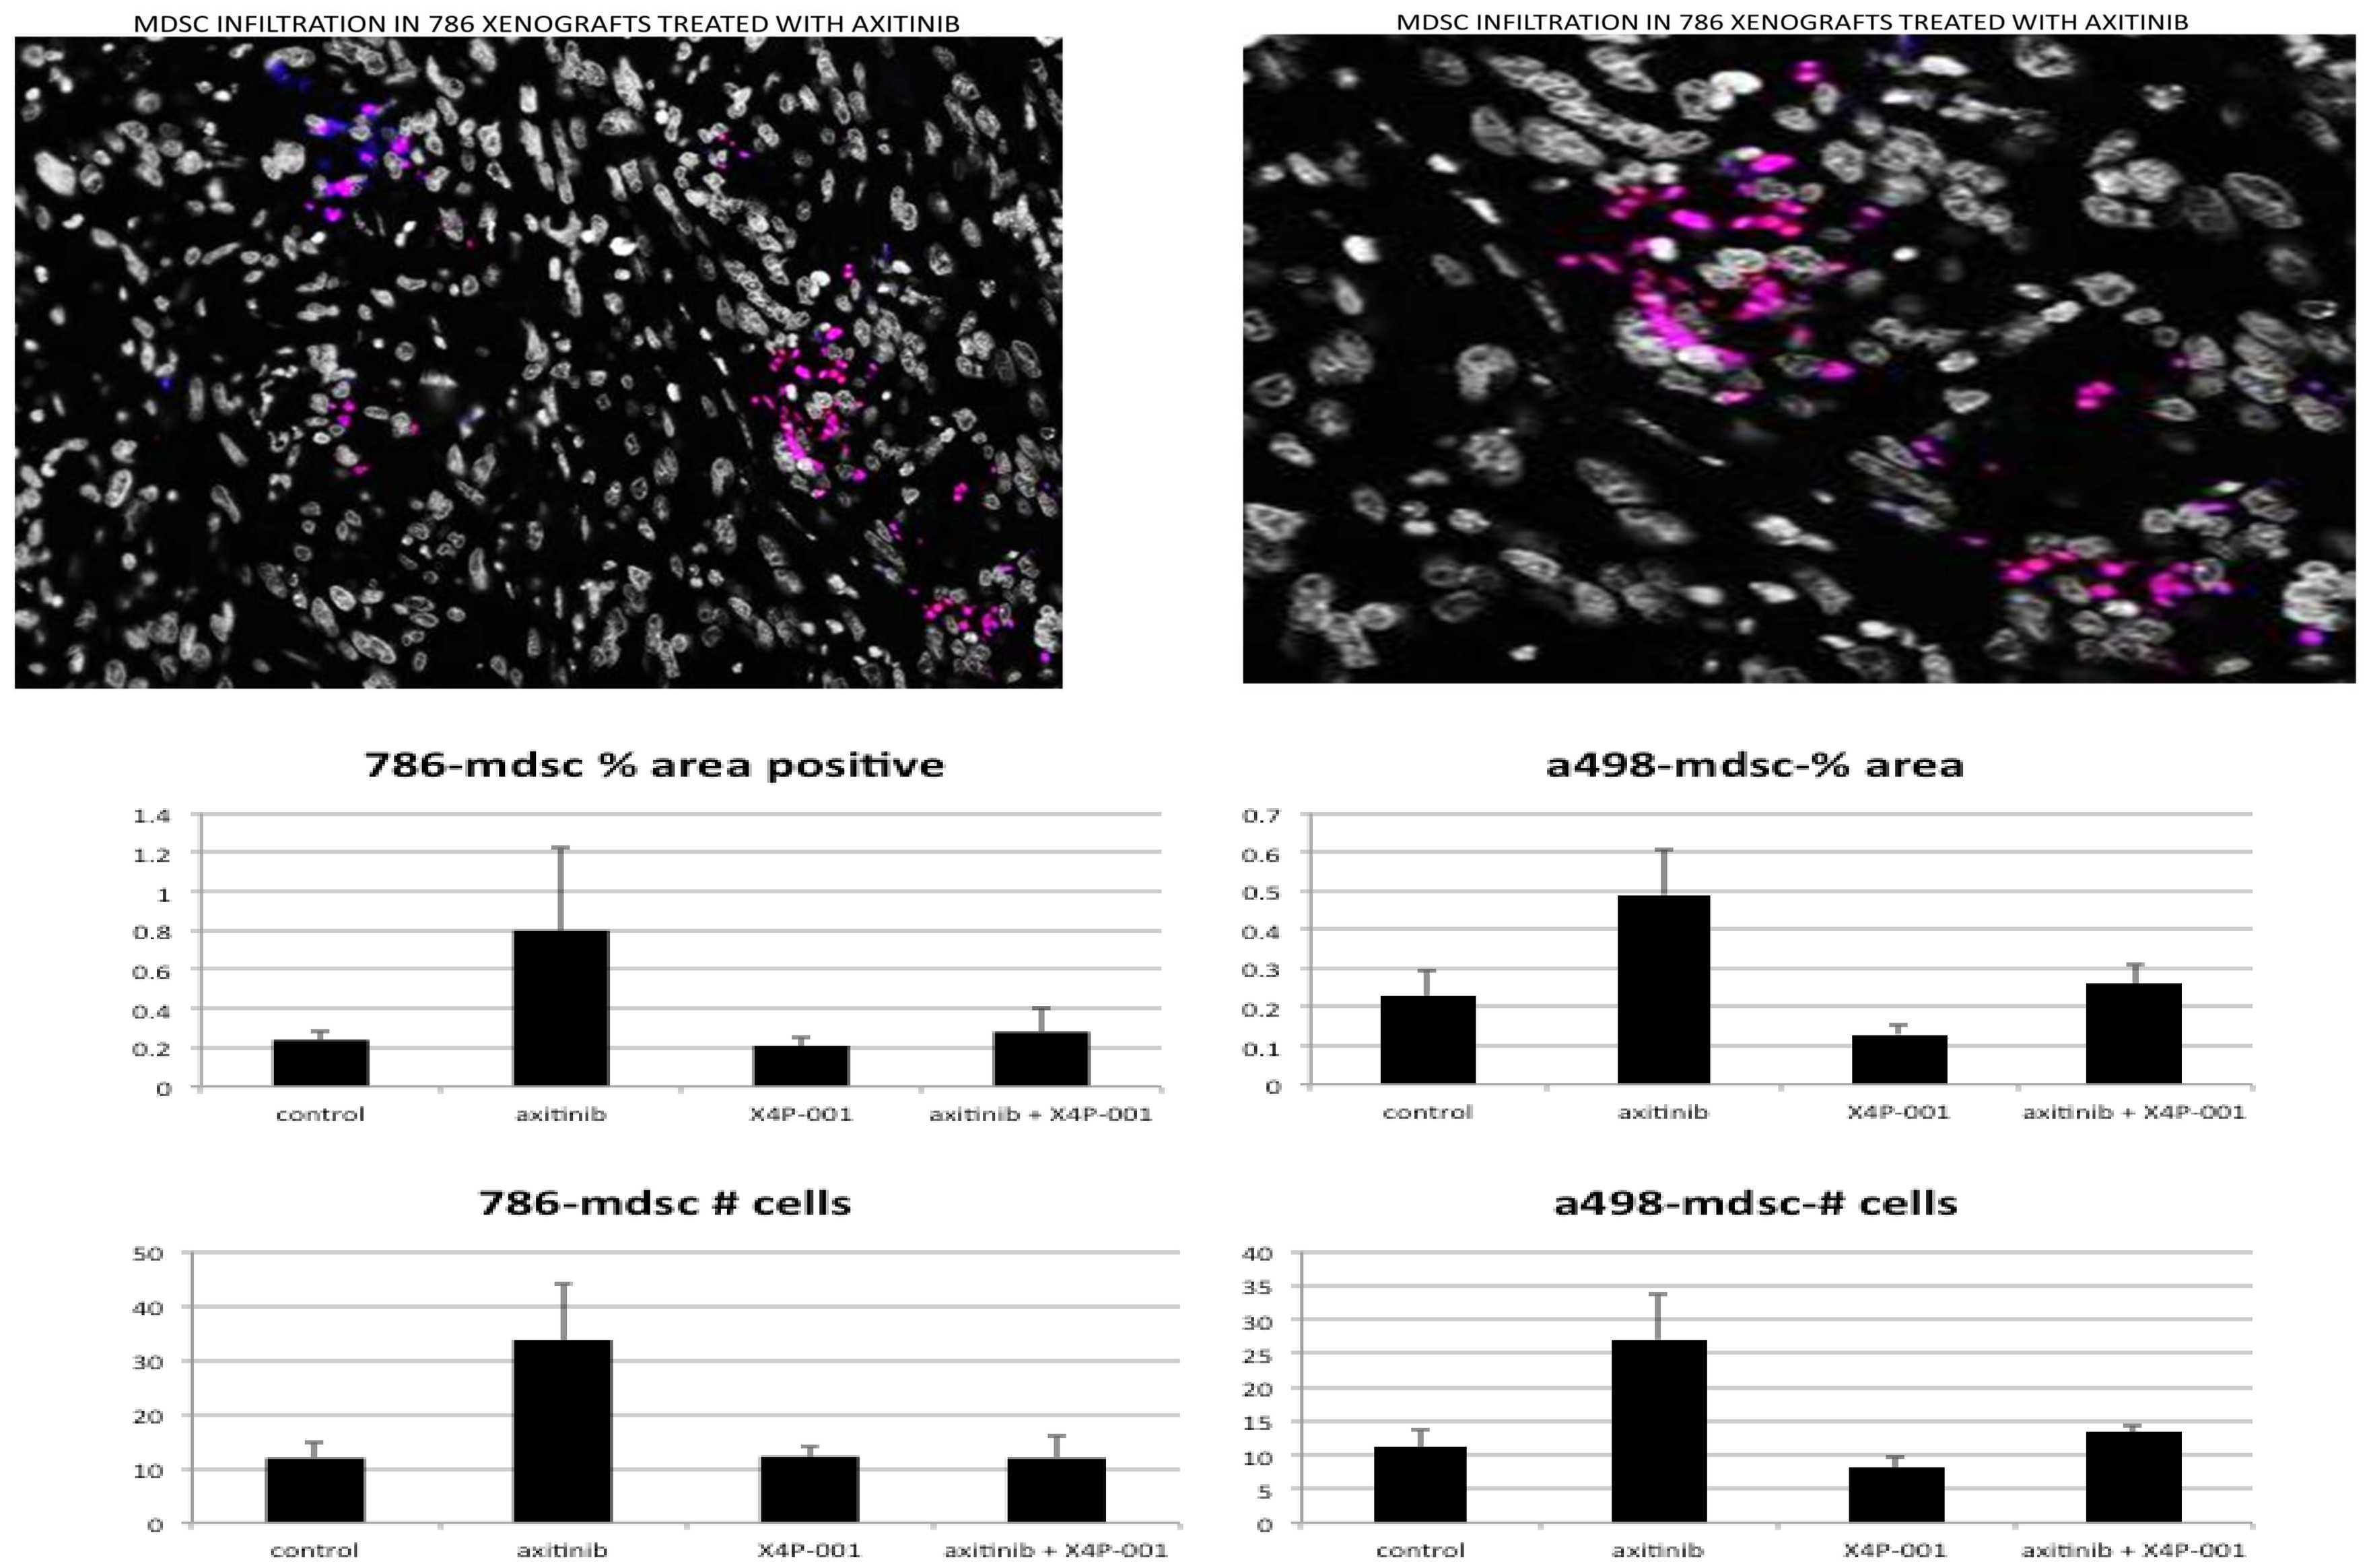
MDSC INFILTRATION IN 786 XENOGRAFTS TREATED WITH AXITINIB
MDSC INFILTRATION IN 786 XENOGRAFTS TREATED WITH AXITINIB
786-mdsc % area positive
a498-mdsc-% area
786-mdsc # cells
a498-mdsc-# cells
control
axitinib
X4P-001
axitinib + X4P-001

Figure 4. Immunofluorescence staining for CD11b (blue) and Gr-1 (red) Positive MDSCs. Top: representative tumor from a single mouse treated with axitinib only. Images were taken at 20X (left) and 40X (right). Bottom: Bar graphs of quantitative analyis of all tumors in each treatment group.

Modulation of key signaling pathways

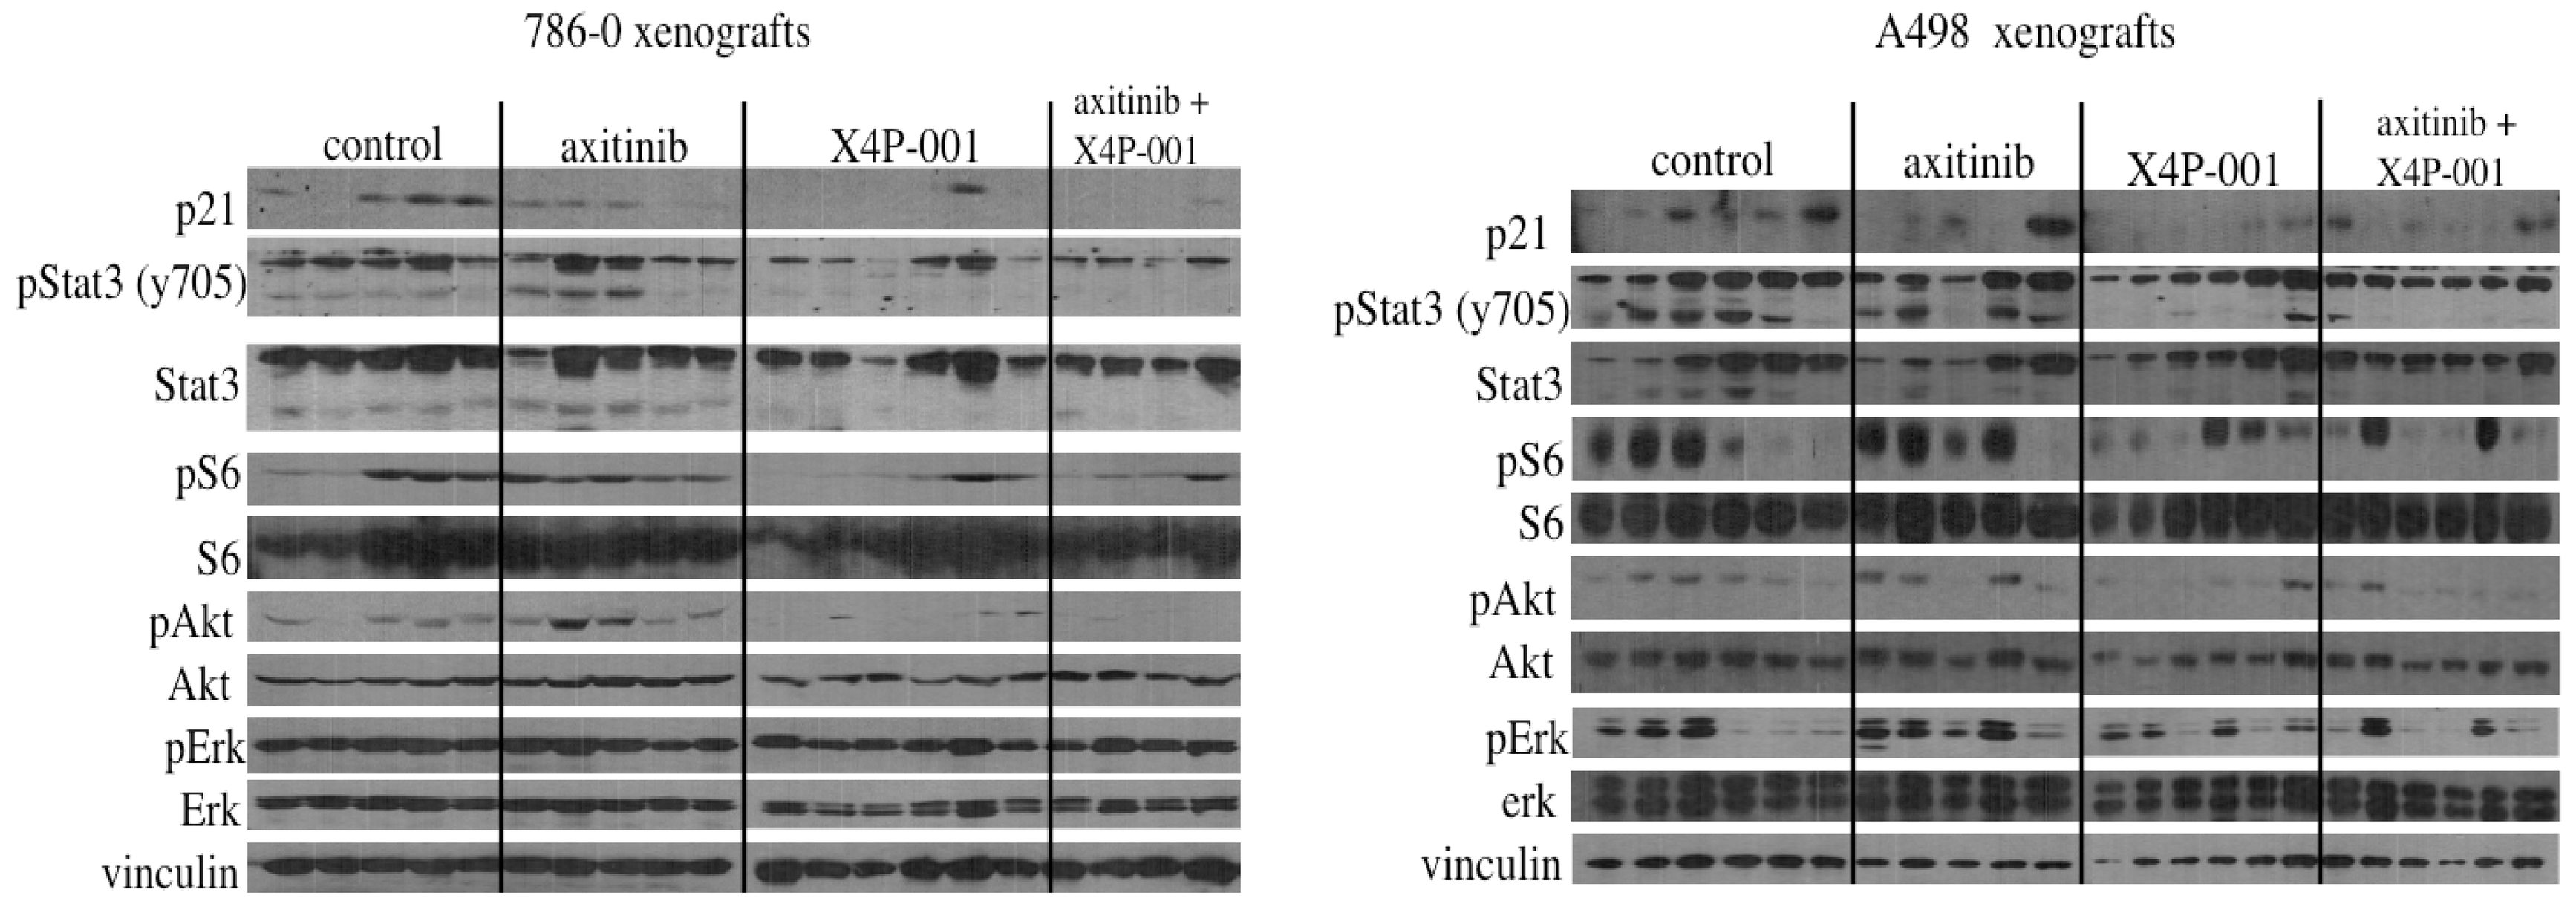
786-0 xenografts
A498 xenografts
control
axitinib
X4P-001
axitinib + X4P-001
p21
pStat3 (y705)
Stat3
pS6
S6
pAkt
Akt
pErk
Erk
erk
vinculin

Figure 5. Western blots of lysates from 786 and A498 xenograft tumors treated with axitinib in the presence and absence of X4P-001

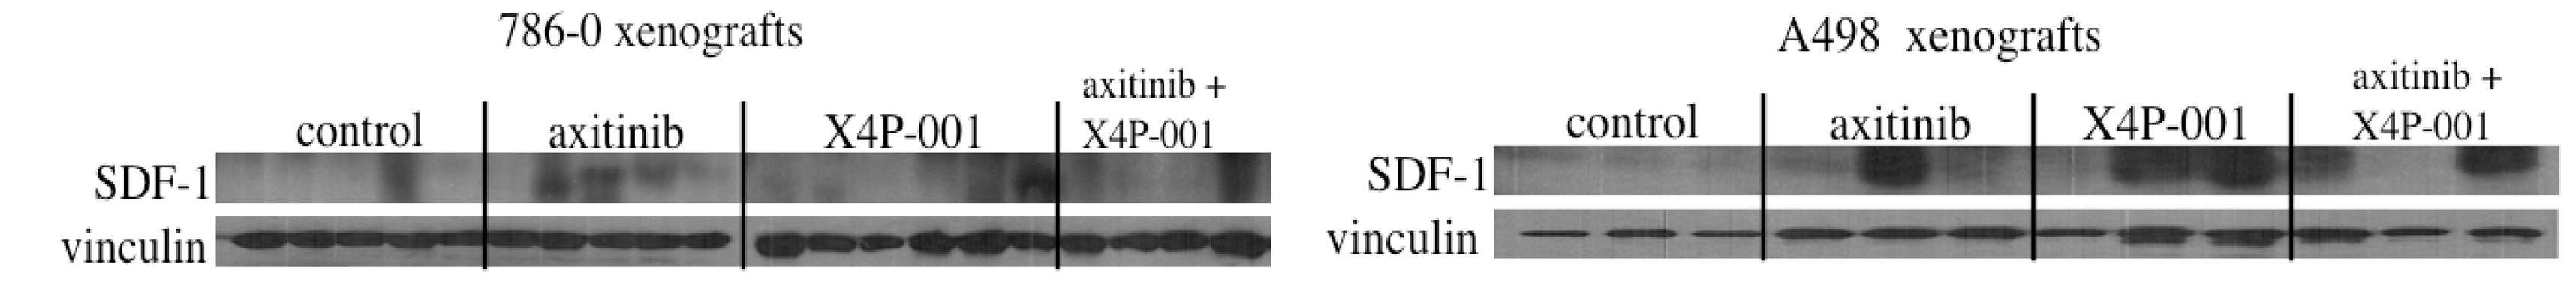
786-0 xenografts
A498 xenografts
control
axitinib
X4P-001
axitinib + X4P-001
SDF-1
vinculin

Figure 6. Western blots of lysates for SDF-1 from 786 and A498 xenograft tumors treated with axitinib in the presence and absence of X4P-001